HE-700 REDUCES THE DEVELOPMENT OF LIVER INFLAMMATION IN OBESE HFD-TREATED LDLR-/-.LEIDEN MICE BY AMELIORATING THE BUILD-UP OF HEPATIC CHOLESTEROL

Andrea Mueller[1], Robert Kleemann[2], Eveline Gart[2], Wim van Duyvenvoorde[2], Lars Verschuren[3], Martien Caspers[3], Ivana Bobeldijk[2], Natascha Krömmelbein[1], Kanita Salic[2], Yvonne Burmeister[1], Bernd Seilheimer[1], Martine C. Morrison[2].

[1] Biologische Heilmittel Heel GmbH, Baden-Baden, Germany.
[2] Department of Metabolic Health Research, The Netherlands Organisation for Applied Scientific Research (TNO) , Leiden, the Netherlands.
[3] Department of Microbiology and Systems Biology, TNO, Zeist, the Netherlands.

INTRODUCTION AND OBJECTIVES

NAFLD is a complex multifactorial disorder that is characterized by dysfunction of hepatic lipid metabolism and chronic inflammation in the liver. NAFLD has become the most common cause of chronic liver disease in many countries, and its prevalence continues to rise in parallel with increasing rates of obesity.

HE-700 is a multicomponent medicinal product that consists of natural ingredients. This product was previously shown to reduce NAFLD activity (NAS) and fibrosis in a study in STAM mice (chemically-induced liver damage combined with HFD). Here we evaluated the potential NAFLD-attenuating effects of HE-700 in a translational diet-induced obese model of NAFLD.

METHODS

Ldlr-/-.Leiden mice, a preclinical model for obesity-associated NASH with translational characteristics (1-3), were fed a HFD for 24 weeks to induce metabolic dysfunction including hepatic steatosis and inflammation.

HE-700 or vehicle control (saline) was administered intraperitoneally 3 x weekly at 1.5 ml/kg from week 6 until the end of the study.

NAFLD/NASH development was assessed histologically (by a board-certified pathologist) and was combined with extensive hepatic gene expression analysis (next generation sequencing), the results of which were substantiated with immunohistochemistry and biochemical analysis of liver lipids.

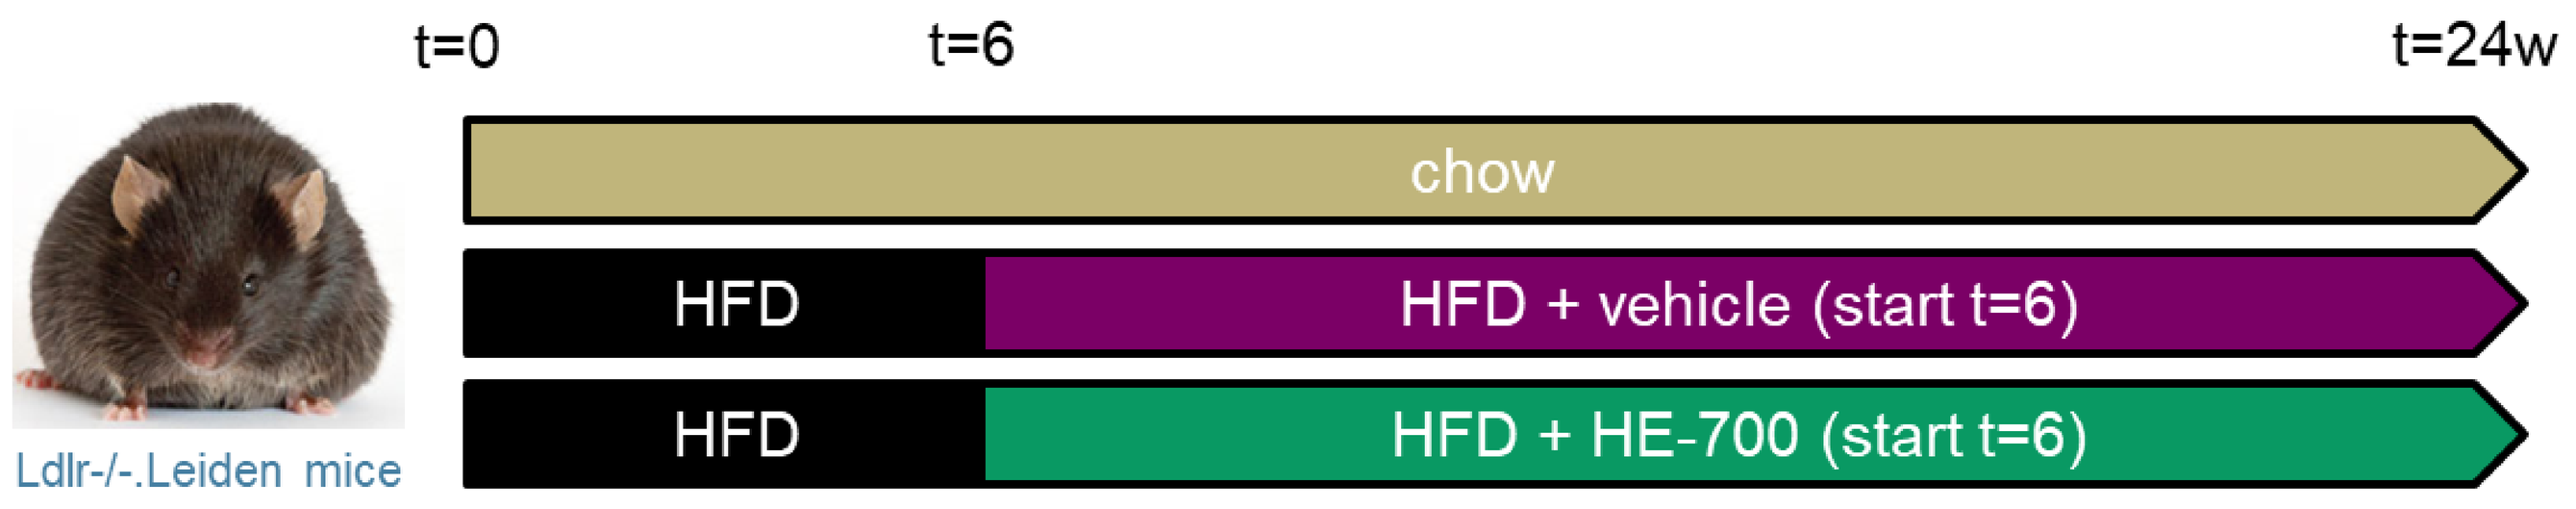
t=0
t=6
t=24w
chow
HFD
HFD + vehicle (start t=6)
HFD
HFD + HE-700 (start t=6)
Ldlr-/-.Leiden mice

HE-700 ATTENUATES DEVELOPMENT OF NASH

HE-700 did not affect body weight, food intake, or metabolic risk factors such as blood glucose, plasma insulin, cholesterol, or triglycerides.

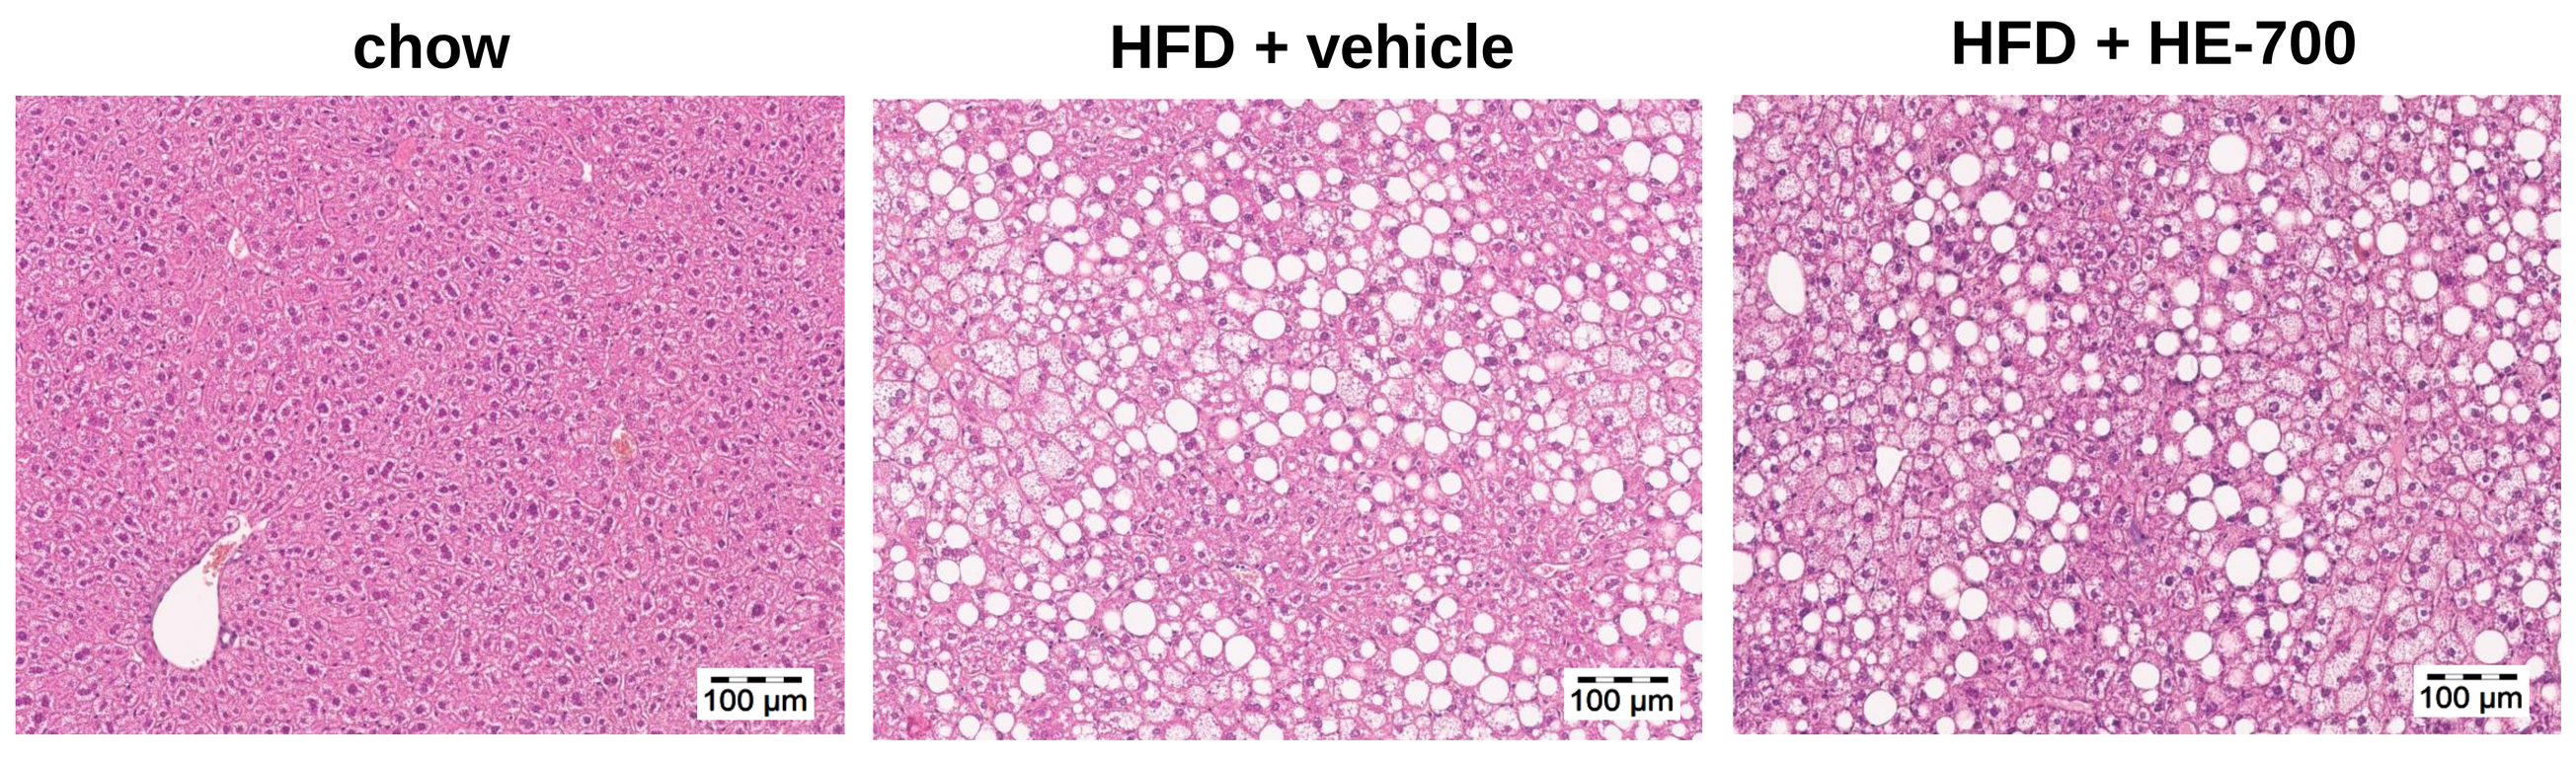
chow
HFD + vehicle
HFD + HE-700
100 µm
100 µm
100 µm

Representative images of HE-stained liver cross sections.

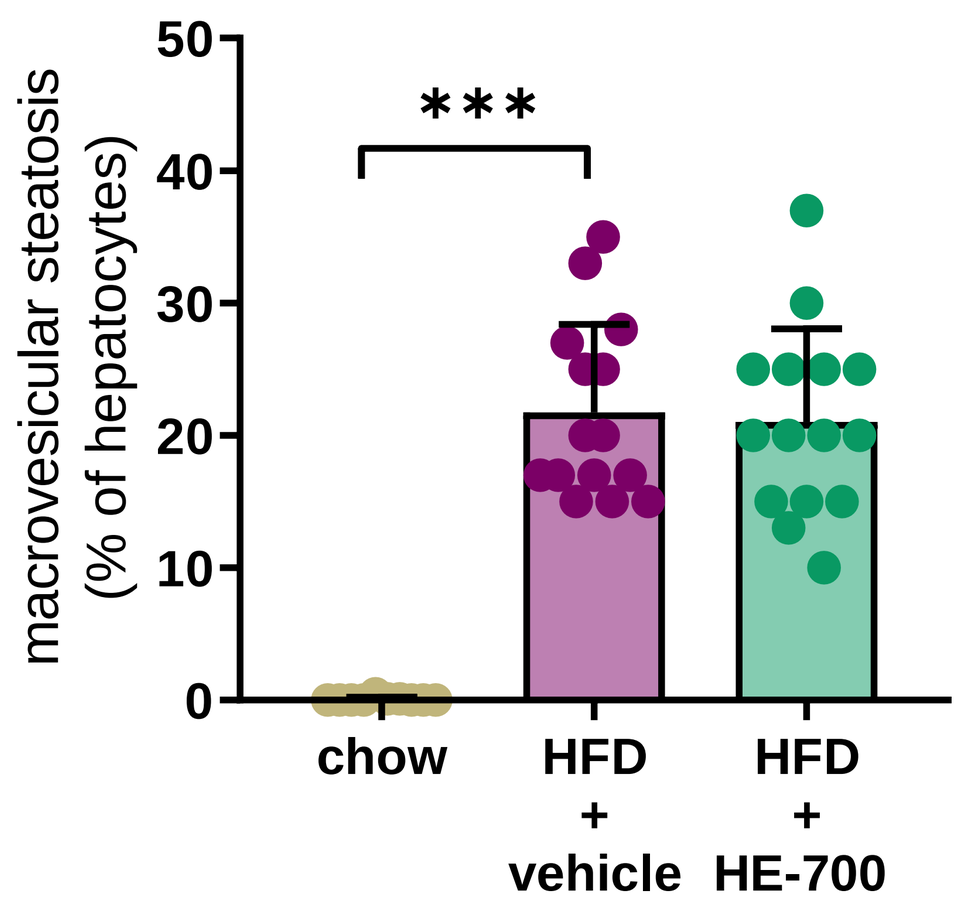
macrovesicular steatosis (% of hepatocytes)
50
40
30
20
10
0

chow
HFD + vehicle
HFD + HE-700

HE-700 did not affect hepatic steatosis.

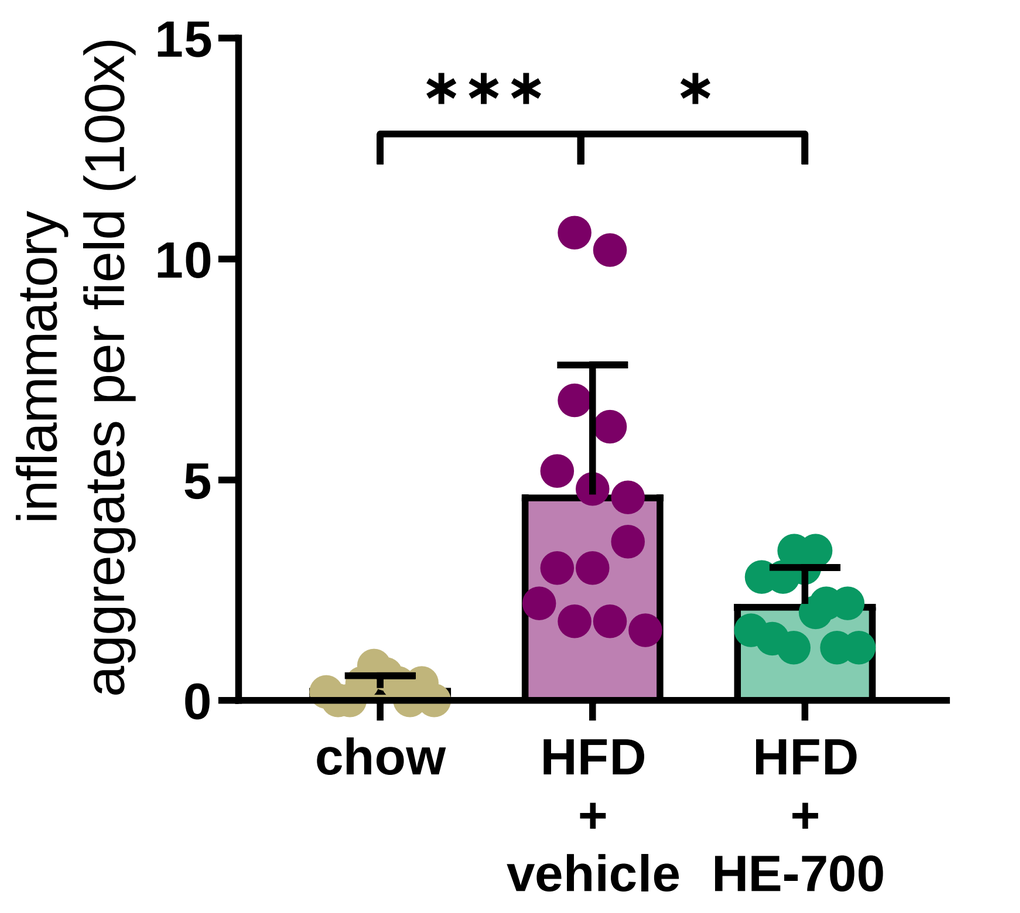
inflammatory aggregates per field (100x)
15
10
5
0

*
chow
HFD + vehicle
HFD + HE-700

HE-700 significantly reduced hepatic inflammation.

Histopathological analysis of NASH showed that HE-700 significantly reduced hepatic inflammation without affecting hepatic steatosis.

martine.morrison@tno.nl

HE-700 MODULATES EXPRESSION OF GENES ASSOCIATED WITH INFLAMMATION AND LIPID METABOLISM

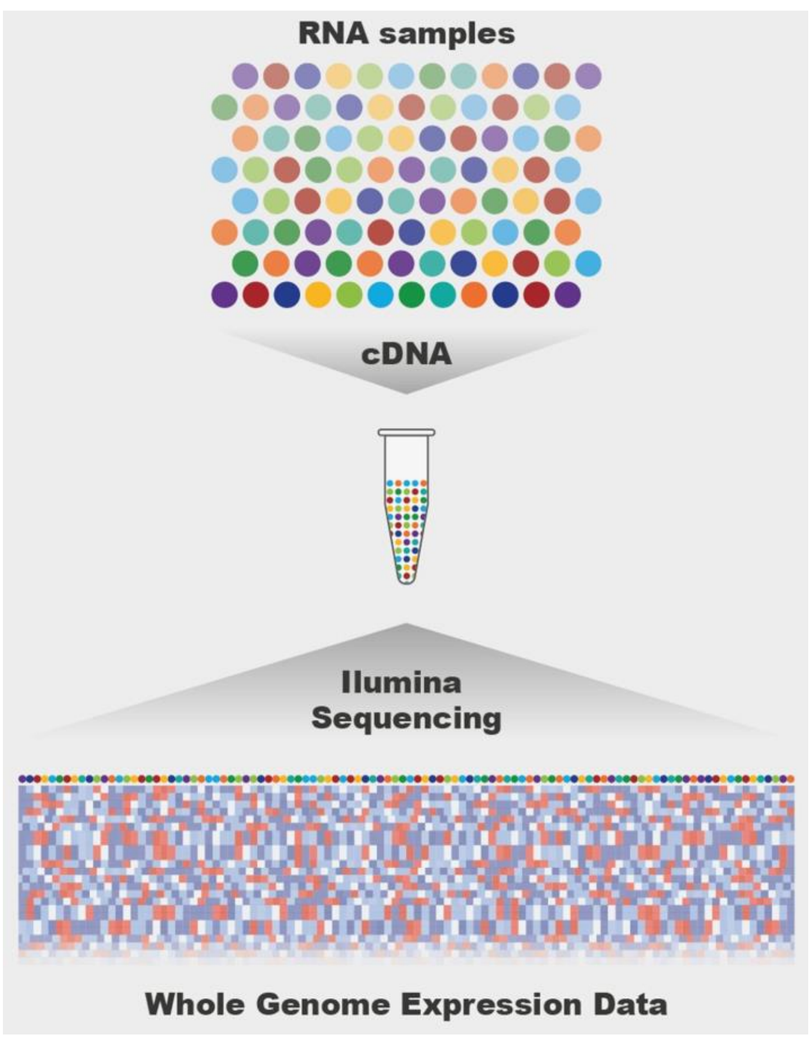
RNA samples
cDNA
Ilumina Sequencing
Whole Genome Expression Data

Hepatic transcriptomics analysis:

- Modulation of inflammatory response (e.g. *Socs3*, *Gzma* and neutrophil chemokine *Cxcl1*).
- Modulation of genes involved in lipid processing (e.g. *Abcg8* and *Pla2g4c*) and upstream regulator cholesterol.

→ Effect of HE-700 on neutrophils?

→ Effect of HE-700 on cholesterol accumulation?

TREATMENT WITH HE-700 REDUCES HEPATIC NEUTROPHIL INFILTRATION

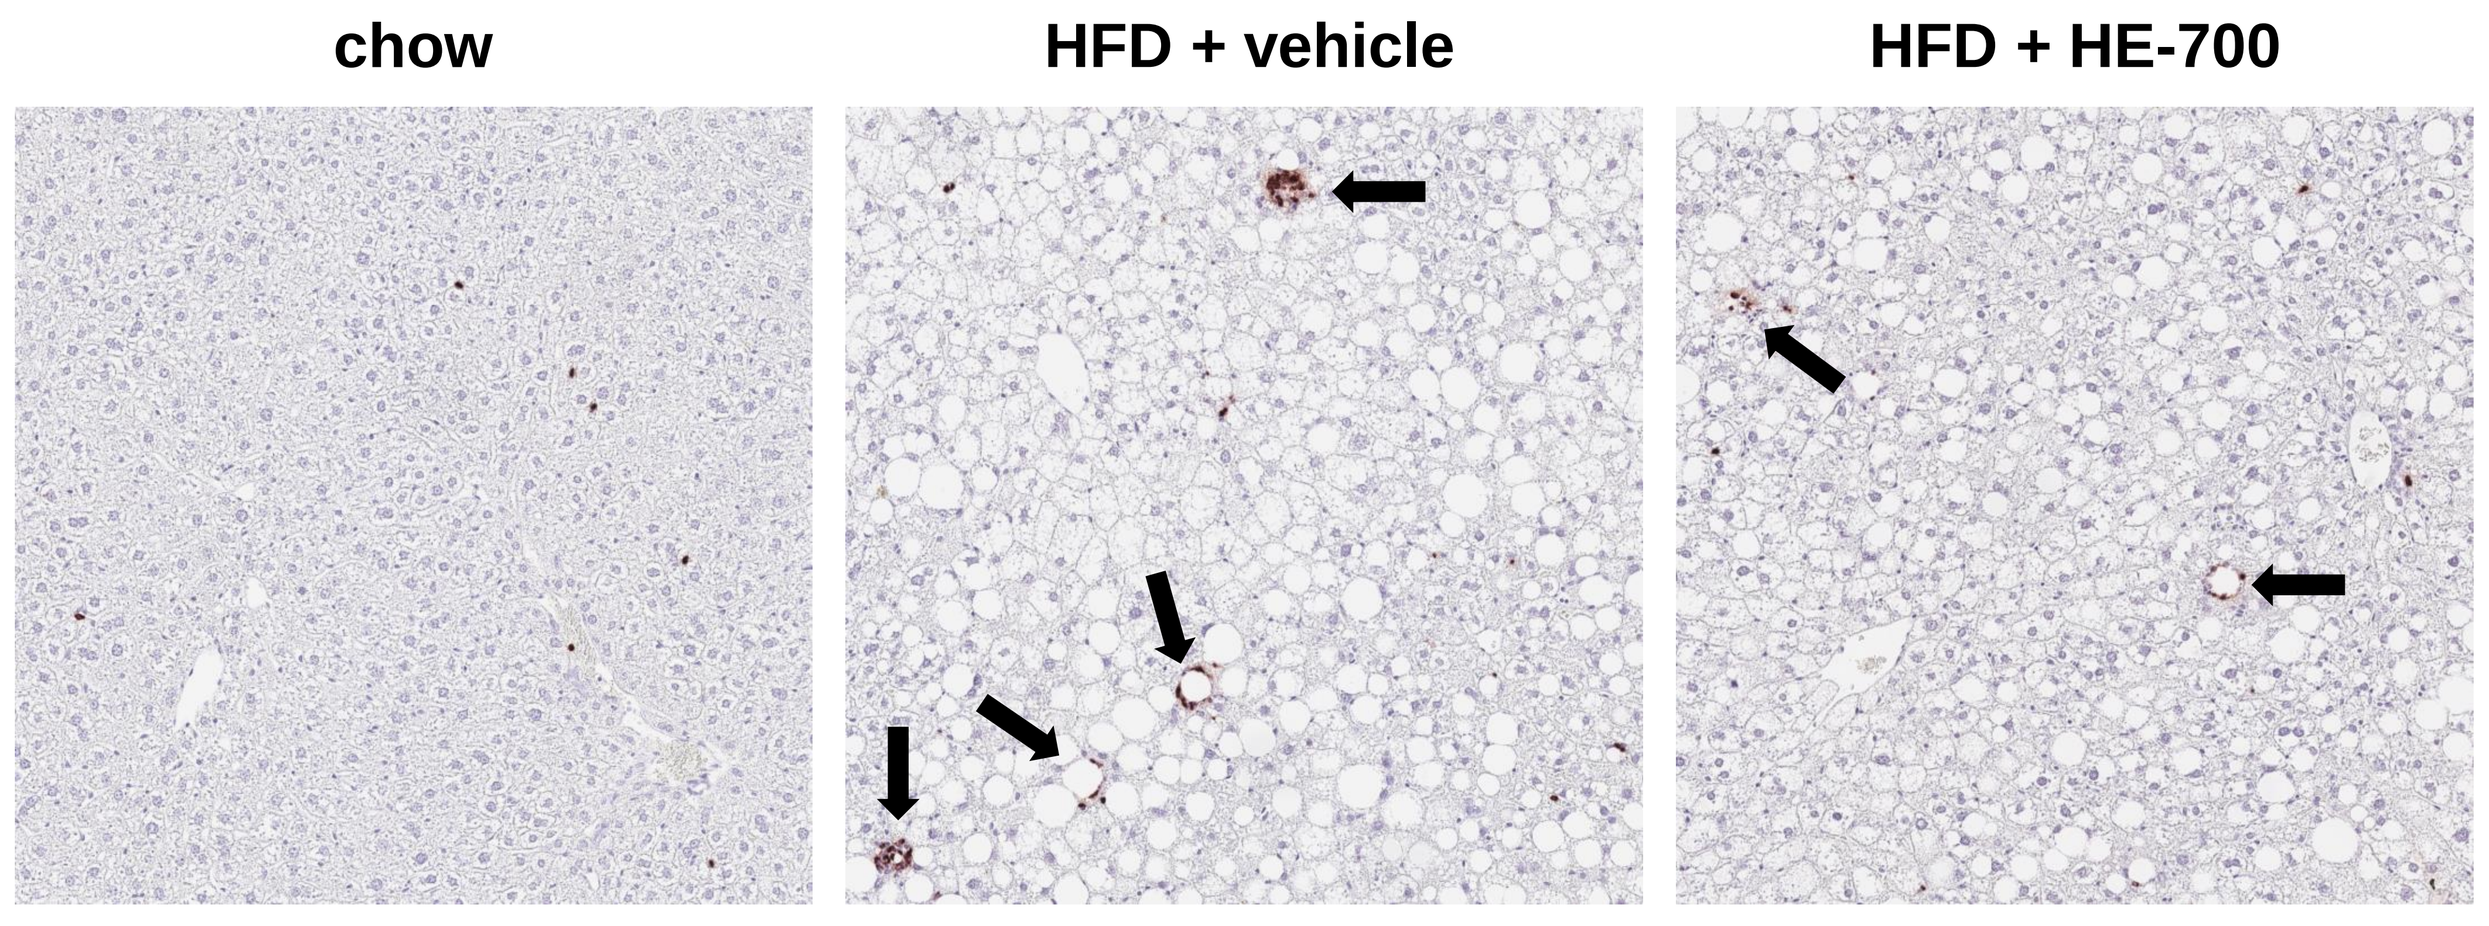
chow
HFD + vehicle
HFD + HE-700

Representative images of GR1-stained liver cross sections. Arrows indicate neutrophil aggregates.

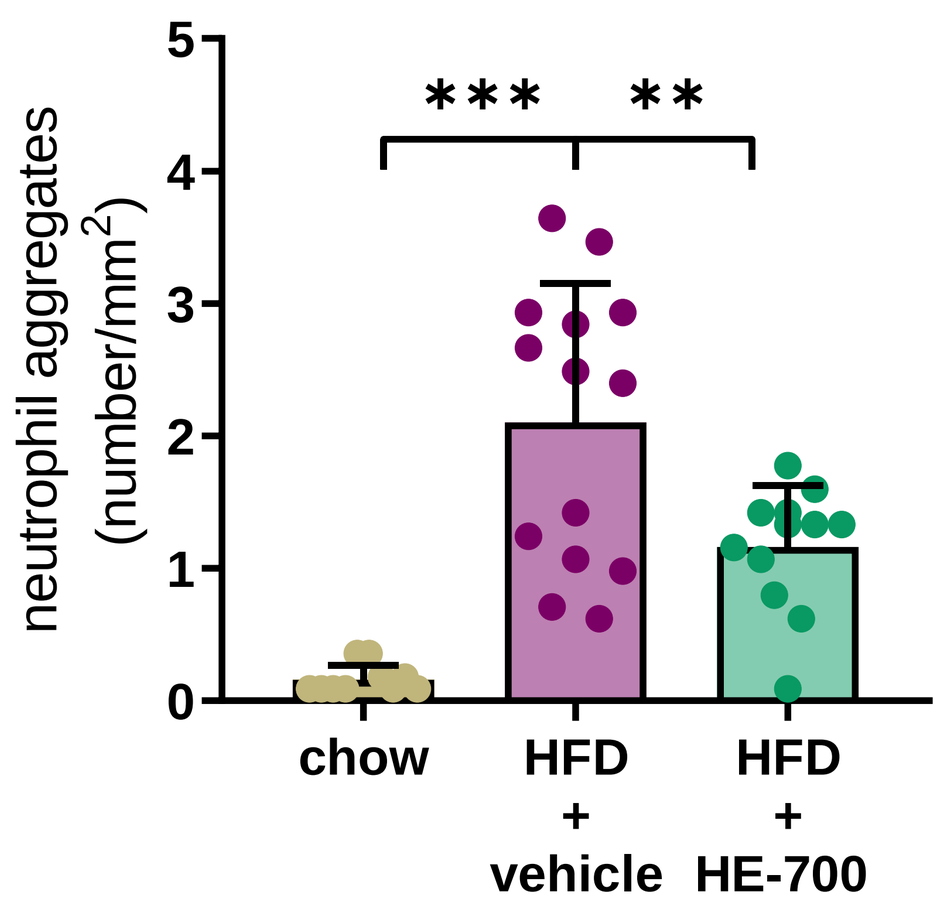
neutrophil aggregates (number/mm²)
5
4
3
2
1
0

**
chow
HFD + vehicle
HFD + HE-700

Quantification of neutrophil clusters in the liver.

Immunohistochemical staining of neutrophils confirmed the effects on the gene expression level, showing reduced infiltration of neutrophils in the liver.

HE-700 REDUCES HEPATIC CHOLESTEROL ACCUMULATION - POTENTIAL RATIONALE FOR ANTI-INFLAMMATORY EFFECTS

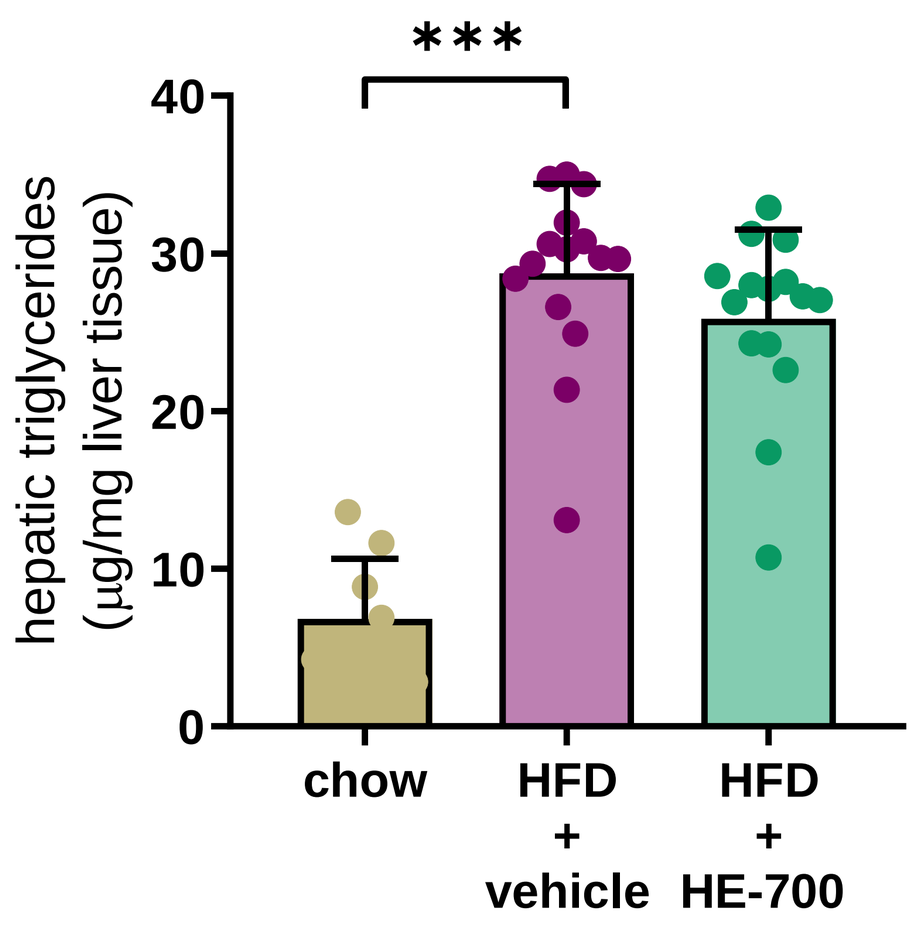
hepatic triglycerides (µg/mg liver tissue)
40
30
20
10
0

chow
HFD + vehicle
HFD + HE-700

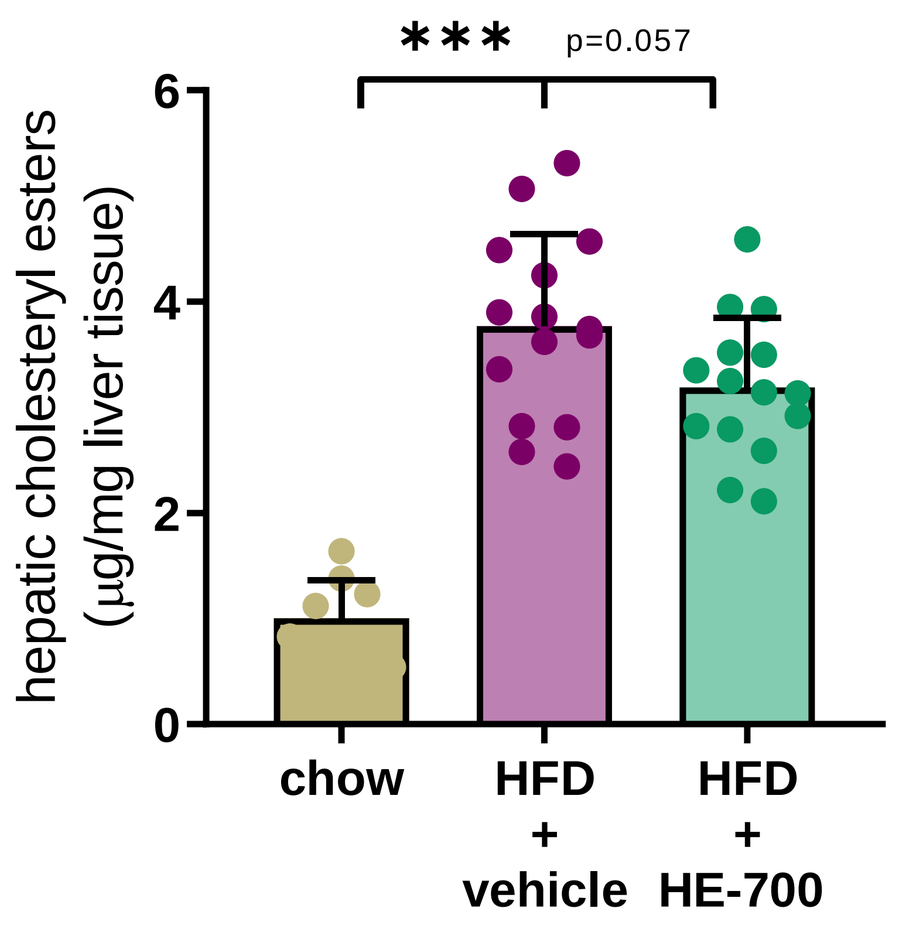
hepatic cholesteryl esters (µg/mg liver tissue)
6
4
2
0

p=0.057
chow
HFD + vehicle
HFD + HE-700

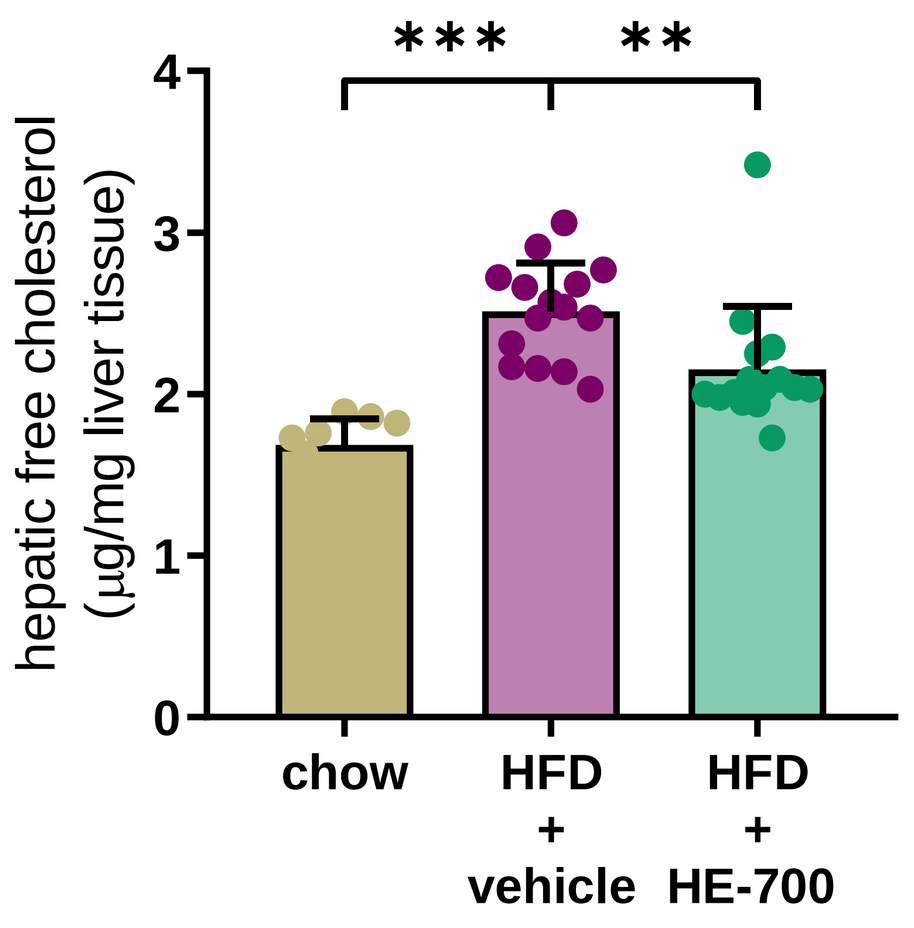
hepatic free cholesterol (µg/mg liver tissue)
4
3
2
1
0

**
chow
HFD + vehicle
HFD + HE-700

Biochemical analysis of intrahepatic lipids showed that HE-700 did not affect the accumulation of triglycerides in the liver but specifically reduced the build-up of (free) cholesterol.

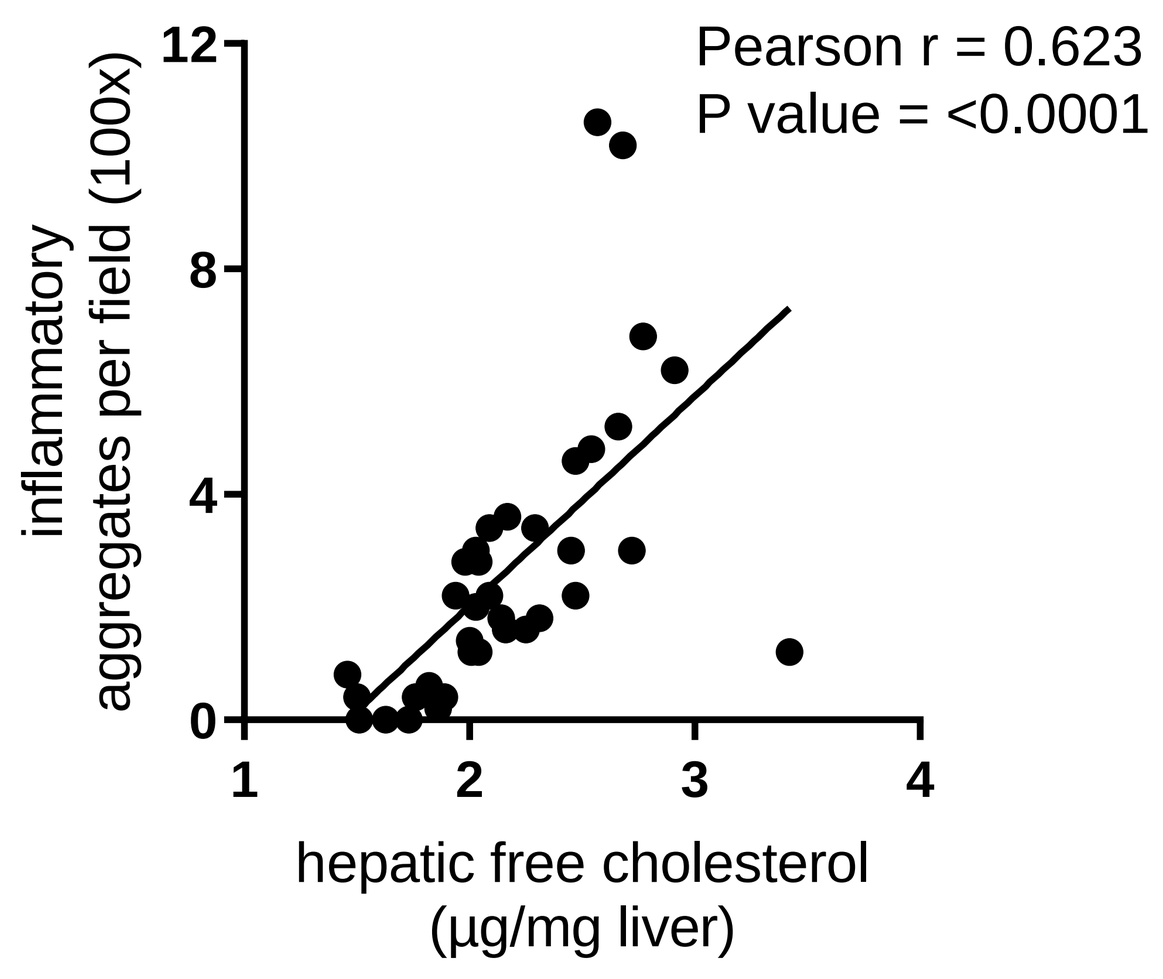
Pearson r = 0.623
P value = <0.0001
inflammatory aggregates per field (100x)
12
8
4
0
1
2
3
4
hepatic free cholesterol (µg/mg liver)

Free cholesterol levels were found to correlate significantly with hepatic inflammatory aggregates, thereby providing a potential rationale for the observed anti-inflammatory effects of HE-700 in the liver.

CONCLUSIONS

HE-700 treatment has anti-inflammatory molecular (e.g. *Cxcl1* expression) and cellular (e.g. neutrophil content) effects in obese Ldlr-/-.Leiden mice with NASH. A reduction of lipotoxic lipid species, i.e. free cholesterol, may provide a rationale for the observed NASH-attenuating effect.

REFERENCES

1. Morrison et al. **Key Inflammatory Processes in Human NASH Are Reflected in Ldlr-/-.Leiden Mice: A Translational Gene Profiling Study. Front Physiol.** *2018 Feb 23;9:132.*

2. Van Koppen et al. **Uncovering a Predictive Molecular Signature for the Onset of NASH-Related Fibrosis in a Translational NASH Mouse Model.** *Cell Mol Gastroenterol Hepatol. 2017 Oct 14;5(1):83-98.e10.*

3. Morrison et al. **Obeticholic Acid Modulates Serum Metabolites and Gene Signatures Characteristic of Human NASH and Attenuates Inflammation and Fibrosis Progression in Ldlr-/-.Leiden Mice.** *Hepatol Commun. 2018 Oct 29;2(12):1513-1532.*